American Society of Clinical Oncology®, June 3-7, 2022

Evaluation of booster injections in maintaining peak immunity in a phase IIb study evaluating HER2/neu peptide GP2 + GM-CSF (GLSI-100) vs. GM-CSF alone after adjuvant trastuzumab in HER2 positive women with breast cancer

Snehal S Patel, David B McWilliams, Mira Patel, Jaye Thompson, F Joseph Daugherty.
Greenwich LifeSciences, Stafford, TX

BACKGROUND

The results of a prospective, randomized, placebo-controlled, single-blinded, multicenter Phase IIb trial investigating GLSI-100 (GP2+GM-CSF) administered in the extended-adjuvant setting to node-positive and high-risk node-negative breast cancer patients with tumors expressing any degree of HER2 (immuno-histochemistry [IHC] 1-3+) (NCT00524277) have been reported. The trial enrolled HLA-A*02 patients randomized to receive GLSI-100 versus GM-CSF alone. It was previously reported that completion of the GLSI-100 Primary Immunization Series (PIS) reduced recurrence rates to 0% over a 5 year follow-up period in HER2 3+ patients, who received a standard course of trastuzumab after surgery. Interim analyses for this trial have been previously reported by Mittendorf et al.

METHODS

Patients were randomly scheduled to receive a total of 6 GLSI-100 (500 mcg GP2: 125 mcg GM-CSF) or GM-CSF only intradermal injections every 3-4 weeks as part of the PIS for the first 6 months and 4 GLSI-100 or GM-CSF only booster intradermal injections every 6 months thereafter. Injection site reactions (ISR) were measured 48-72 hours after injection. The largest perpendicular diameters of induration were measured, and the orthogonal mean was calculated to quantify ISRs. These assessments allow some measure of peak and nadir immunity during the PIS and booster phases of treatment.

Enroll

Baseline DTH

Randomize

GP2 + GM-CSF

GM-CSF alone

6 Month DTH after PIS

Primary Objective
- Determine if GP2+GM-CSF treatment reduces recurrence rates vs. GM-CSF alone

Secondary Objective
- Monitor immune response and correlate with clinical outcomes
- Monitor for any unexpected toxicities

RESULTS

Figure 1: After 5 years of follow-up, the Kaplan-Meier estimated that the 5-year DFS rate in the 46 HER2 3+ patients treated with GLSI-100, if the patients were treated, followed, and remained disease free over the first six months of treatment, was 100% versus 89.4% (95% CI:76.2, 95.5%) in the 50 placebo patients treated with GM-CSF (p = 0.0338). Figure 1 describes the median orthogonal mean of the ISRs over time. Each dose of GLSI-100 generated a significantly larger ISR than the control group. Note that patients who experienced ISRs larger than 100mm had the subsequent dose of GM-CSF reduced, which could have caused the slight reduction in ISRs thereafter.

Figure 1: Disease Free Survival and Injection Site Reactions in HER2 3+ (Positive) Population

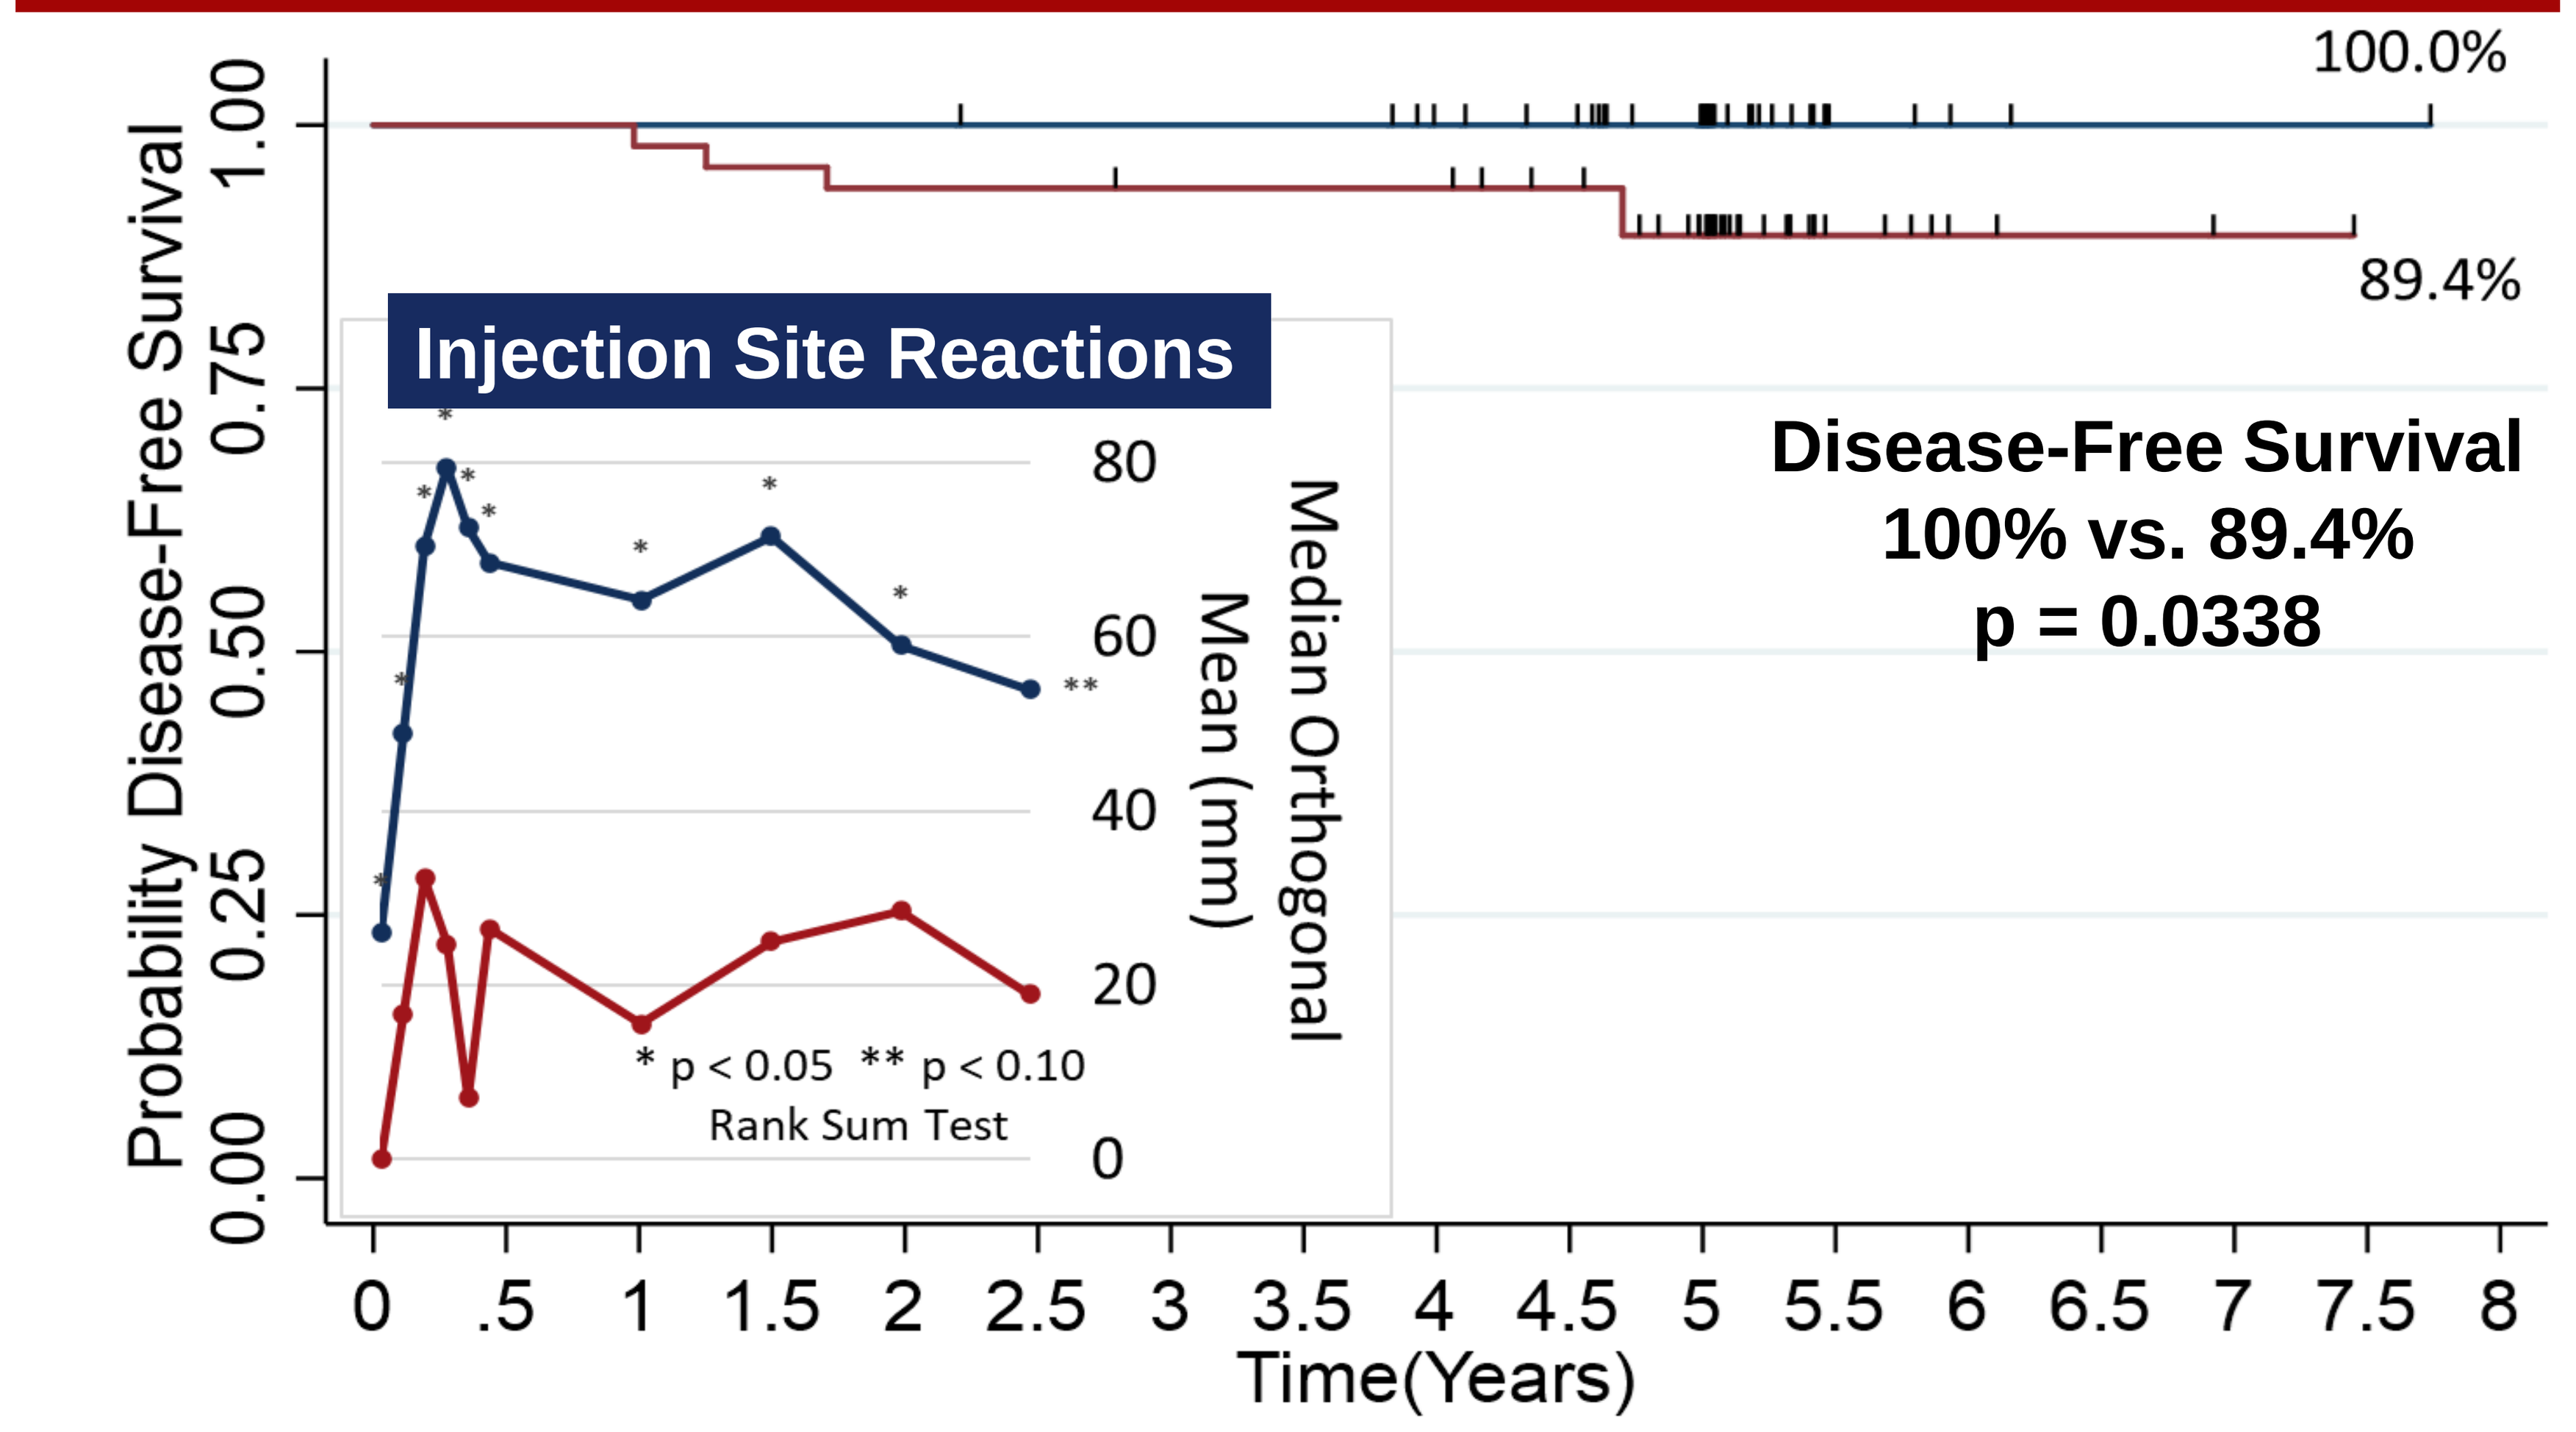
Probability Disease-Free Survival
0.00
0.25
0.50
0.75
1.00
100.0%
89.4%
Injection Site Reactions
80
60
40
20
0
Median Orthogonal Mean (mm)
* p < 0.05 ** p < 0.10
Rank Sum Test
Disease-Free Survival
100% vs. 89.4%
p = 0.0338
0 .5 1 1.5 2 2.5 3 3.5 4 4.5 5 5.5 6 6.5 7 7.5 8
Time(Years)

GP2	46	46	46	46	46	45	45	45	42	40	30	4	2	1	1	1	0
Placebo	50	50	49	48	47	47	46	46	46	43	33	7	3	2	1	0	0

GP2 — Placebo

Figure 2: Percentage of Patients with Maximum Injection Site Reaction at Each PIS Dose in HER2 3+ (Positive) Population – Doses 1-6

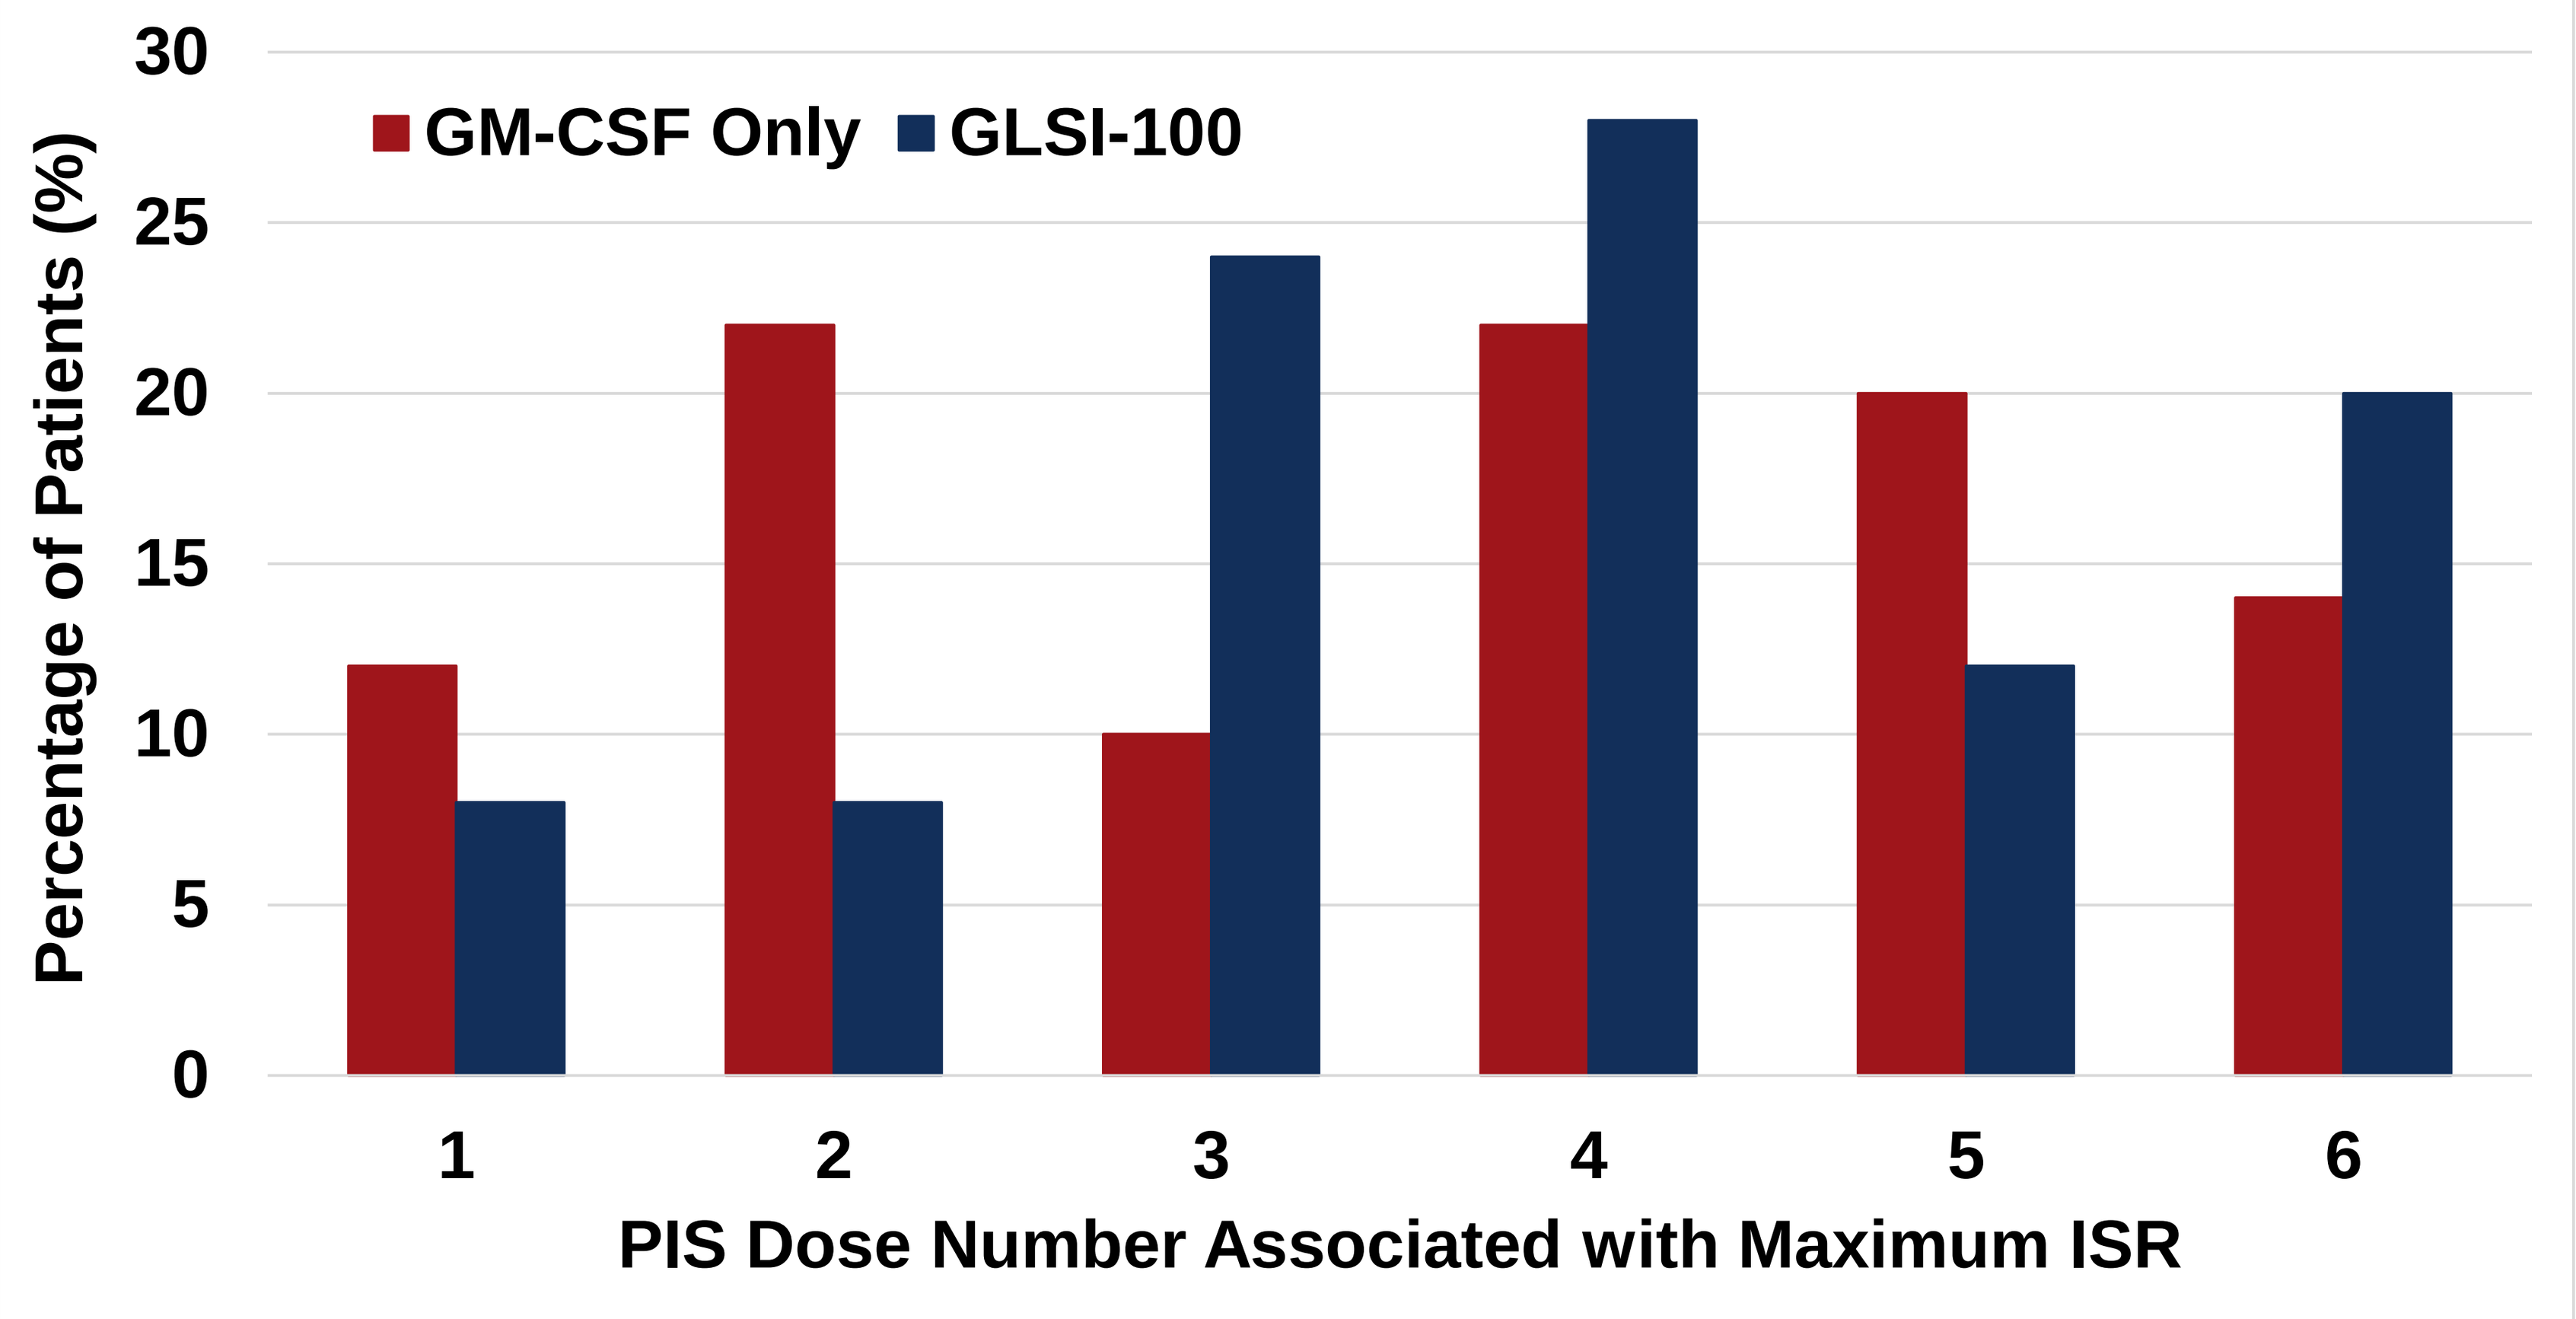
Percentage of Patients (%)
30
25
20
15
10
5
0
GM-CSF Only
GLSI-100
1
2
3
4
5
6
PIS Dose Number Associated with Maximum ISR

Figure 2: The median peak ISR during the PIS in GLSI-100 treated patients was 92.1 mm versus 60.5 mm in GM-CSF control patients. Peak ISR in GLSI-100 patients occurred throughout the 10-dose period. The peak ISR during the PIS dosing period occurred between doses 3 and 6, with 84% of patients experiencing their peak ISR after one of those doses.

Figure 3: Sustainability of Immune Response During Booster Series in HER2 3+ Population – Doses 7-10

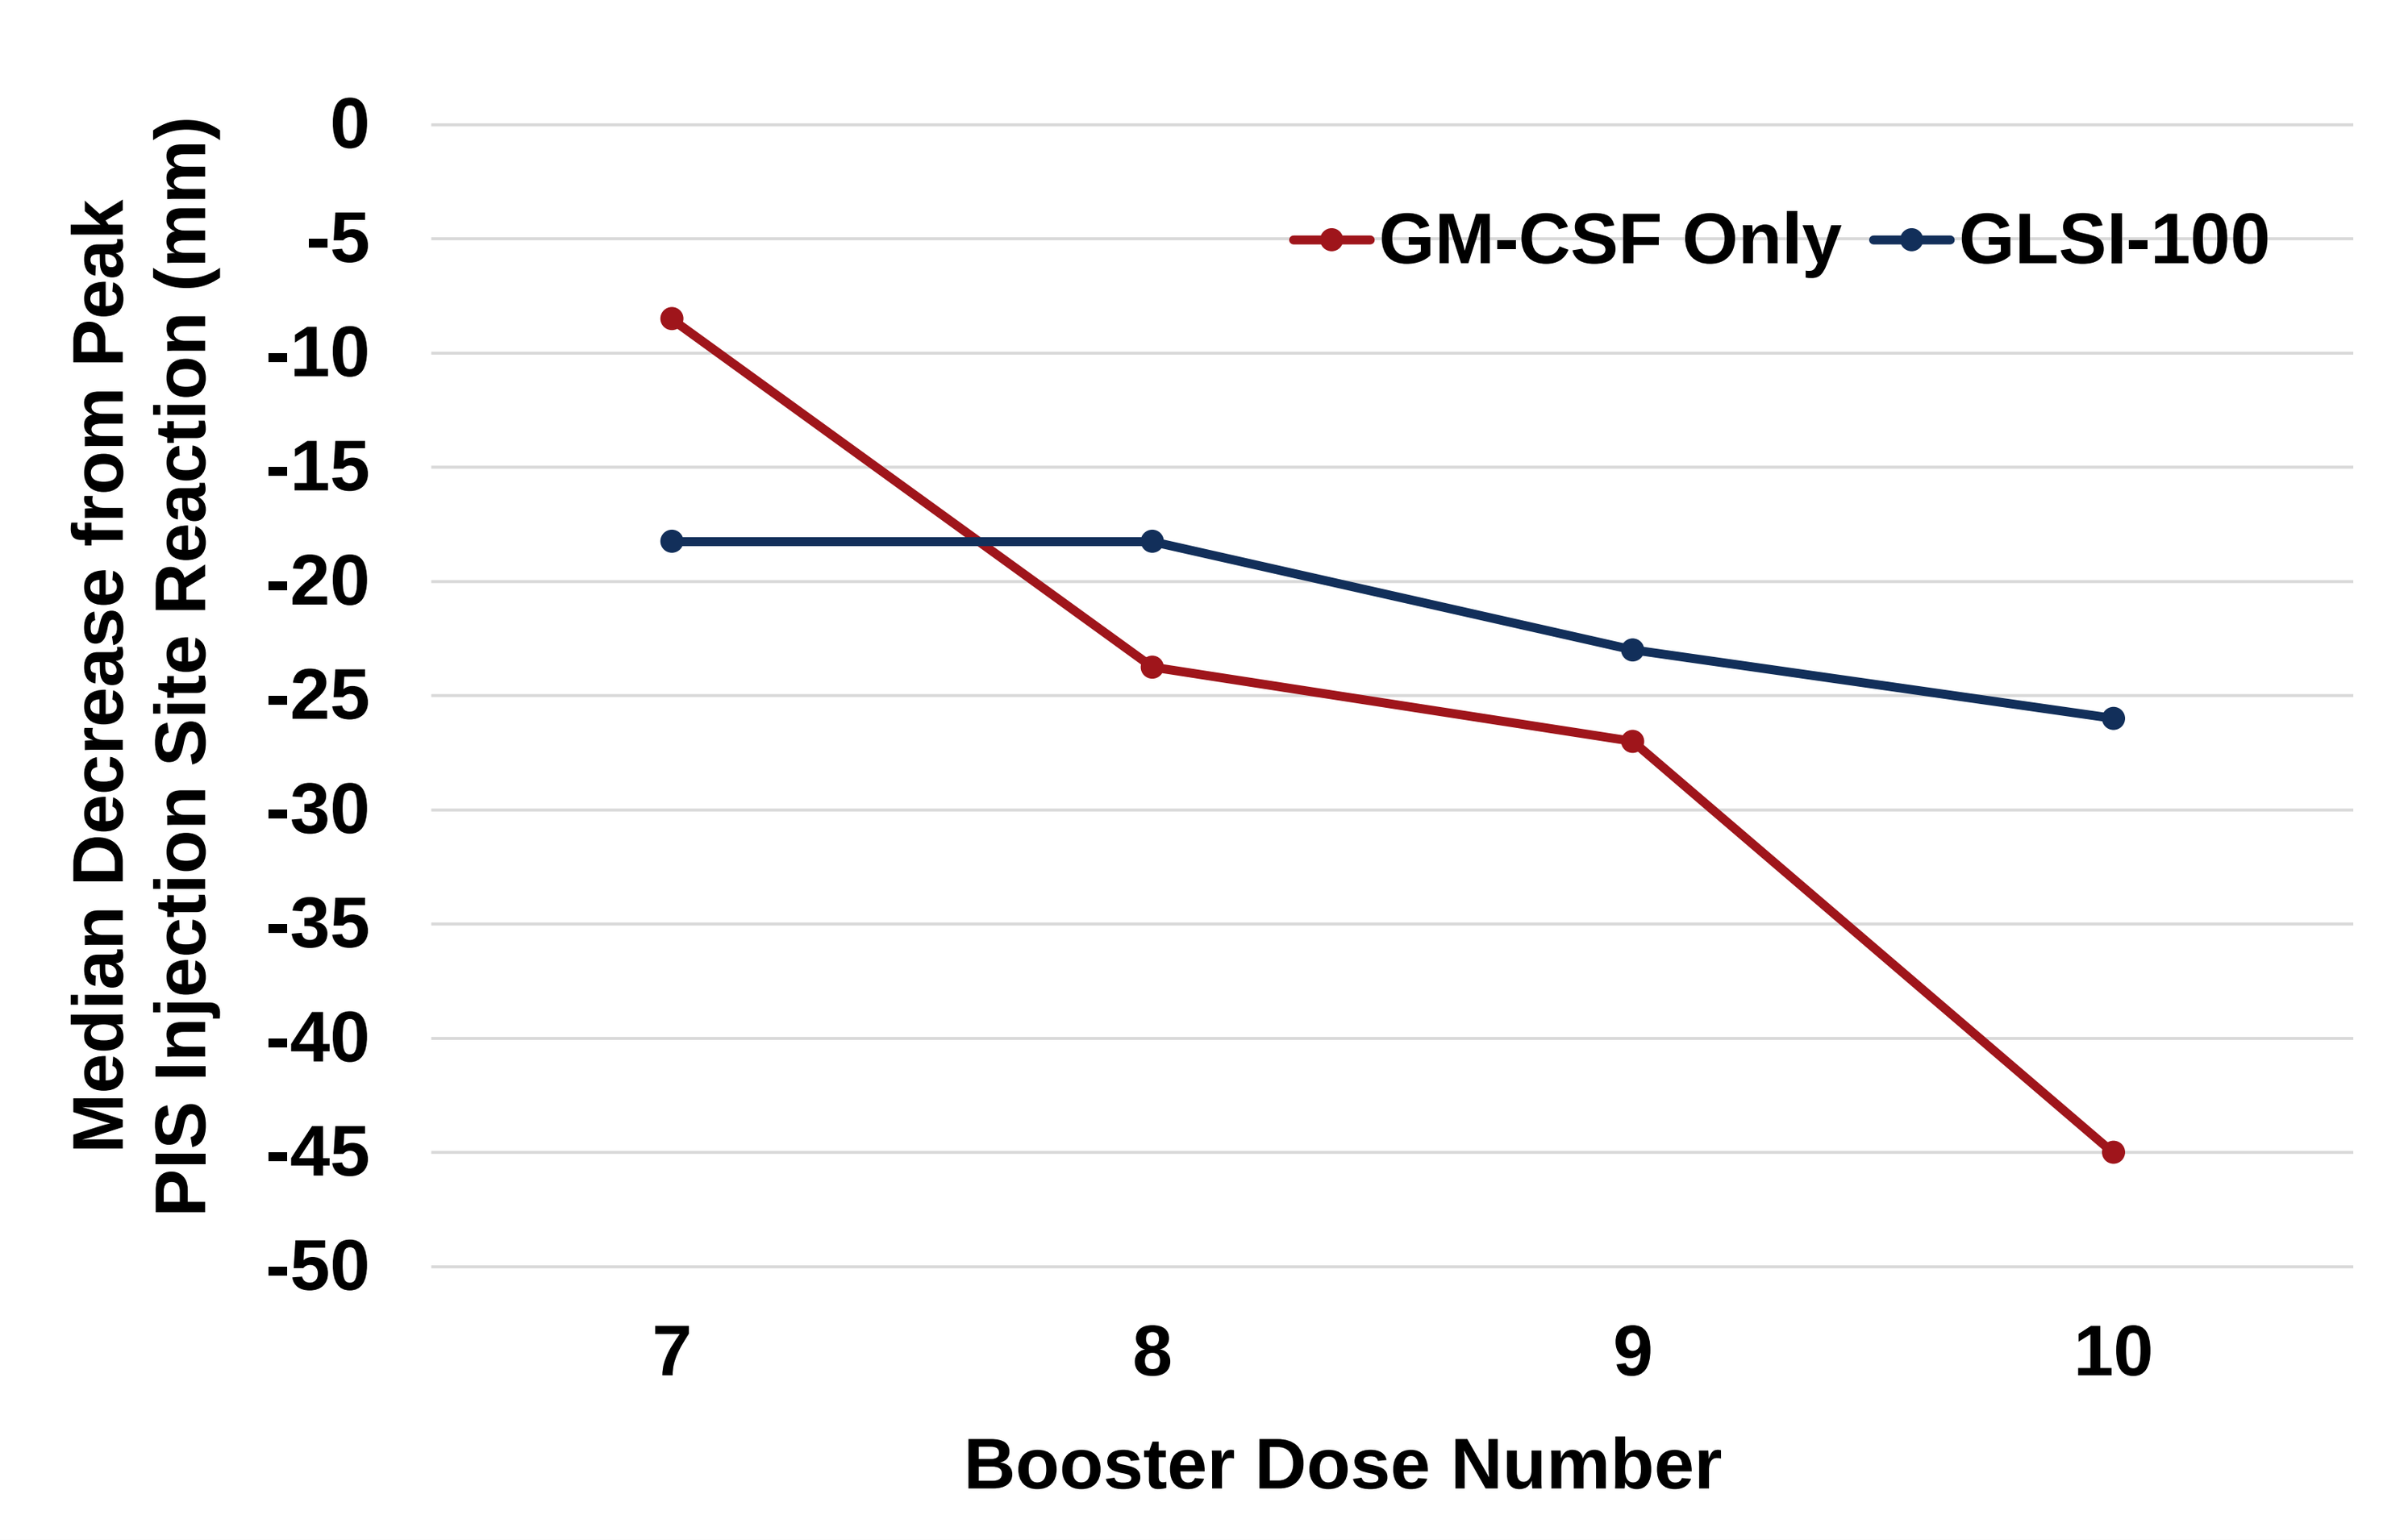
Median Decrease from Peak PIS Injection Site Reaction (mm)
0
-5
-10
-15
-20
-25
-30
-35
-40
-45
-50
GM-CSF Only
GLSI-100
7
8
9
10
Booster Dose Number

Figure 3: Booster doses are administered 6 months after the prior dose. Therefore, ISRs of booster injections may represent a nadir in immune response. The peak ISR orthogonal mean during the PIS was compared to each ISR associated with the booster injections. Booster ISRs in GLSI-100 treated patients were approximately 20mm smaller than the peak ISR during the PIS. However, even with the 20mm reduction, booster ISRs of treated patients were still larger than the average booster ISR of control patients.

DISCUSSION AND CONCLUSIONS

Administering GLSI-100 boosters at 6 month intervals to patients produced a consistent nadir ISR approximately 20 mm lower than the maximum PIS ISR of 92.1 mm, which is still larger than the maximum ISR in GM-CSF only patients of 60.5 mm. A patient's immune response a month after booster dosing would theoretically be the peak ISR, which will be measured in future trials by measuring T-cell response and DTH one month after booster injections, further helping to evaluate booster strategies to sustain peak immunity over longer periods of time.

ACKNOWLEDGEMENTS

The authors have an ownership interest in Greenwich LifeSciences.

This presentation is the intellectual property of the authors/presenters. Contact them at snehal.patel@greenwichlifesciences.com for permission to reprint and/or distribute.

322